P4-10-16

INX-315, an Oral, Potent and Selective CDK2 Inhibitor in Patients with CDK4/6 Inhibitor Resistant ER+/HER2- Breast Cancer or CCNE1 Amplified Solid Tumors:

PHASE 1 MONOTHERAPY DOSE ESCALATION

Antoinette R. Tan, MD, MHSc[1]; Antonio Giordano, MD, PhD[2]; Catherine Shannon, MD[3]; Carey Anders, MD[4]; Nashat Gabrail, MD[5]; Kevin Kalinsky, MD, MS[6] ; Patrick Roberts, PharmD, PhD[7]; Andrew Beelen, MD[7]; George Au-Yeung, MD, PhD[8]

[1]Atrium Health Levine Cancer Institute, Wake Forest University School of Medicine, [2]Dana-Farber Cancer Institute, [3]Mater Hospital, [4]Duke Cancer Center, [5]Gabrail Cancer Center Research, [6]Emory Winship Cancer Institute, [7]Incyclix Bio, LLC, [8]Peter MacCallum Cancer Center

BACKGROUND

- Cyclin-dependent kinases (CDK) are a family of serine / threonine kinases that heterodimerize with regulatory subunits called cyclins to drive cell cycle progression, cell division, and associated biological processes
- CDK2 plays a prominent role in promoting G1/S transition and S phase progression
- Dysregulated CDK2 activity commonly occurs through (i) amplification of CCNE1 (gene that encodes cyclin E1 protein), a known resistance mechanism to oral CDK4/6 inhibitors[1] and/or (ii) overexpression of cyclin E1
- CCNE1 amplified or cyclin E overexpressing cancers have worse survival in several tumor types including ovarian[2,3] and breast cancer[4,5]
- INX-315 is an oral, potent, and selective small molecule inhibitor of CDK2 for the treatment of human cancers[6]

CONCLUSIONS

- INX-315 monotherapy was generally well tolerated with only 1 DLT observed to date
- The majority of TRAEs were grade 1 or 2 with only 16% of patients experiencing a grade 3 or greater TRAE
- INX-315 demonstrated monotherapy antitumor activity in heavily pretreated patients with metastatic ER+/HER2- breast cancer and CCNE1 amplified, metastatic, platinum-resistant ovarian cancer
- Monotherapy and combination dose expansion are ongoing

References:
(1) Jhaveri K, et al. Expert Rev Anticancer Ther. 2021; 21:1105-1124
(2) Kang EY, et al. Cancer. 2023; 129:697-713
(3) Petersen S, et al. Gynecologic oncology. 2020; 157:405-10
(4) Luhtala S, et al. Tumor Biology. 2016; 37:9813-23
(5) Lundgren C, et al. Acta Oncologica. 2015; 54:543-9
(6) Dietrich et al. Cancer Discov. 2024; 14(3):446–467

SAN ANTONIO BREAST CANCER SYMPOSIUM®
DECEMBER 10-13, 2024

METHODS

STUDY DESIGN AND TREATMENT

- INX-315-01 (NCT05735080) is a first-in-human, Phase 1/2, open-label, dose escalation, combination, and dose-expansion study of orally administered INX-315 (Figure 1)

KEY INCLUSION CRITERIA (PART A)

- ER+/HER2- breast cancer that has progressed on/after a CDK4/6 inhibitor
- Platinum-resistant or platinum-refractory CCNE1 amplified (by NGS) epithelial ovarian cancer that has progressed after standard systemic therapy
- Solid tumor with known amplification of CCNE1 (by NGS) that has progressed after standard therapy, been intolerant to or is ineligible for standard therapy
- At least 1 measurable lesion per RECIST v1.1
- Age ≥18 years; Eastern Cooperative Oncology Group performance status 0 or 1; and adequate bone marrow, renal, and liver function
- No prior therapy with a CDK2 inhibitor, including CDK2/4/6 inhibitors

ASSESSMENTS (PART A)

- Primary Objectives: safety, tolerability, dose-limiting toxicities, and recommended doses for expansion (RDEs) as monotherapy and in combination with fulvestrant
- Secondary Objectives: characterize the single and multiple dose pharmacokinetics of INX-315 and assess preliminary antitumor activity

STUDY DESIGN

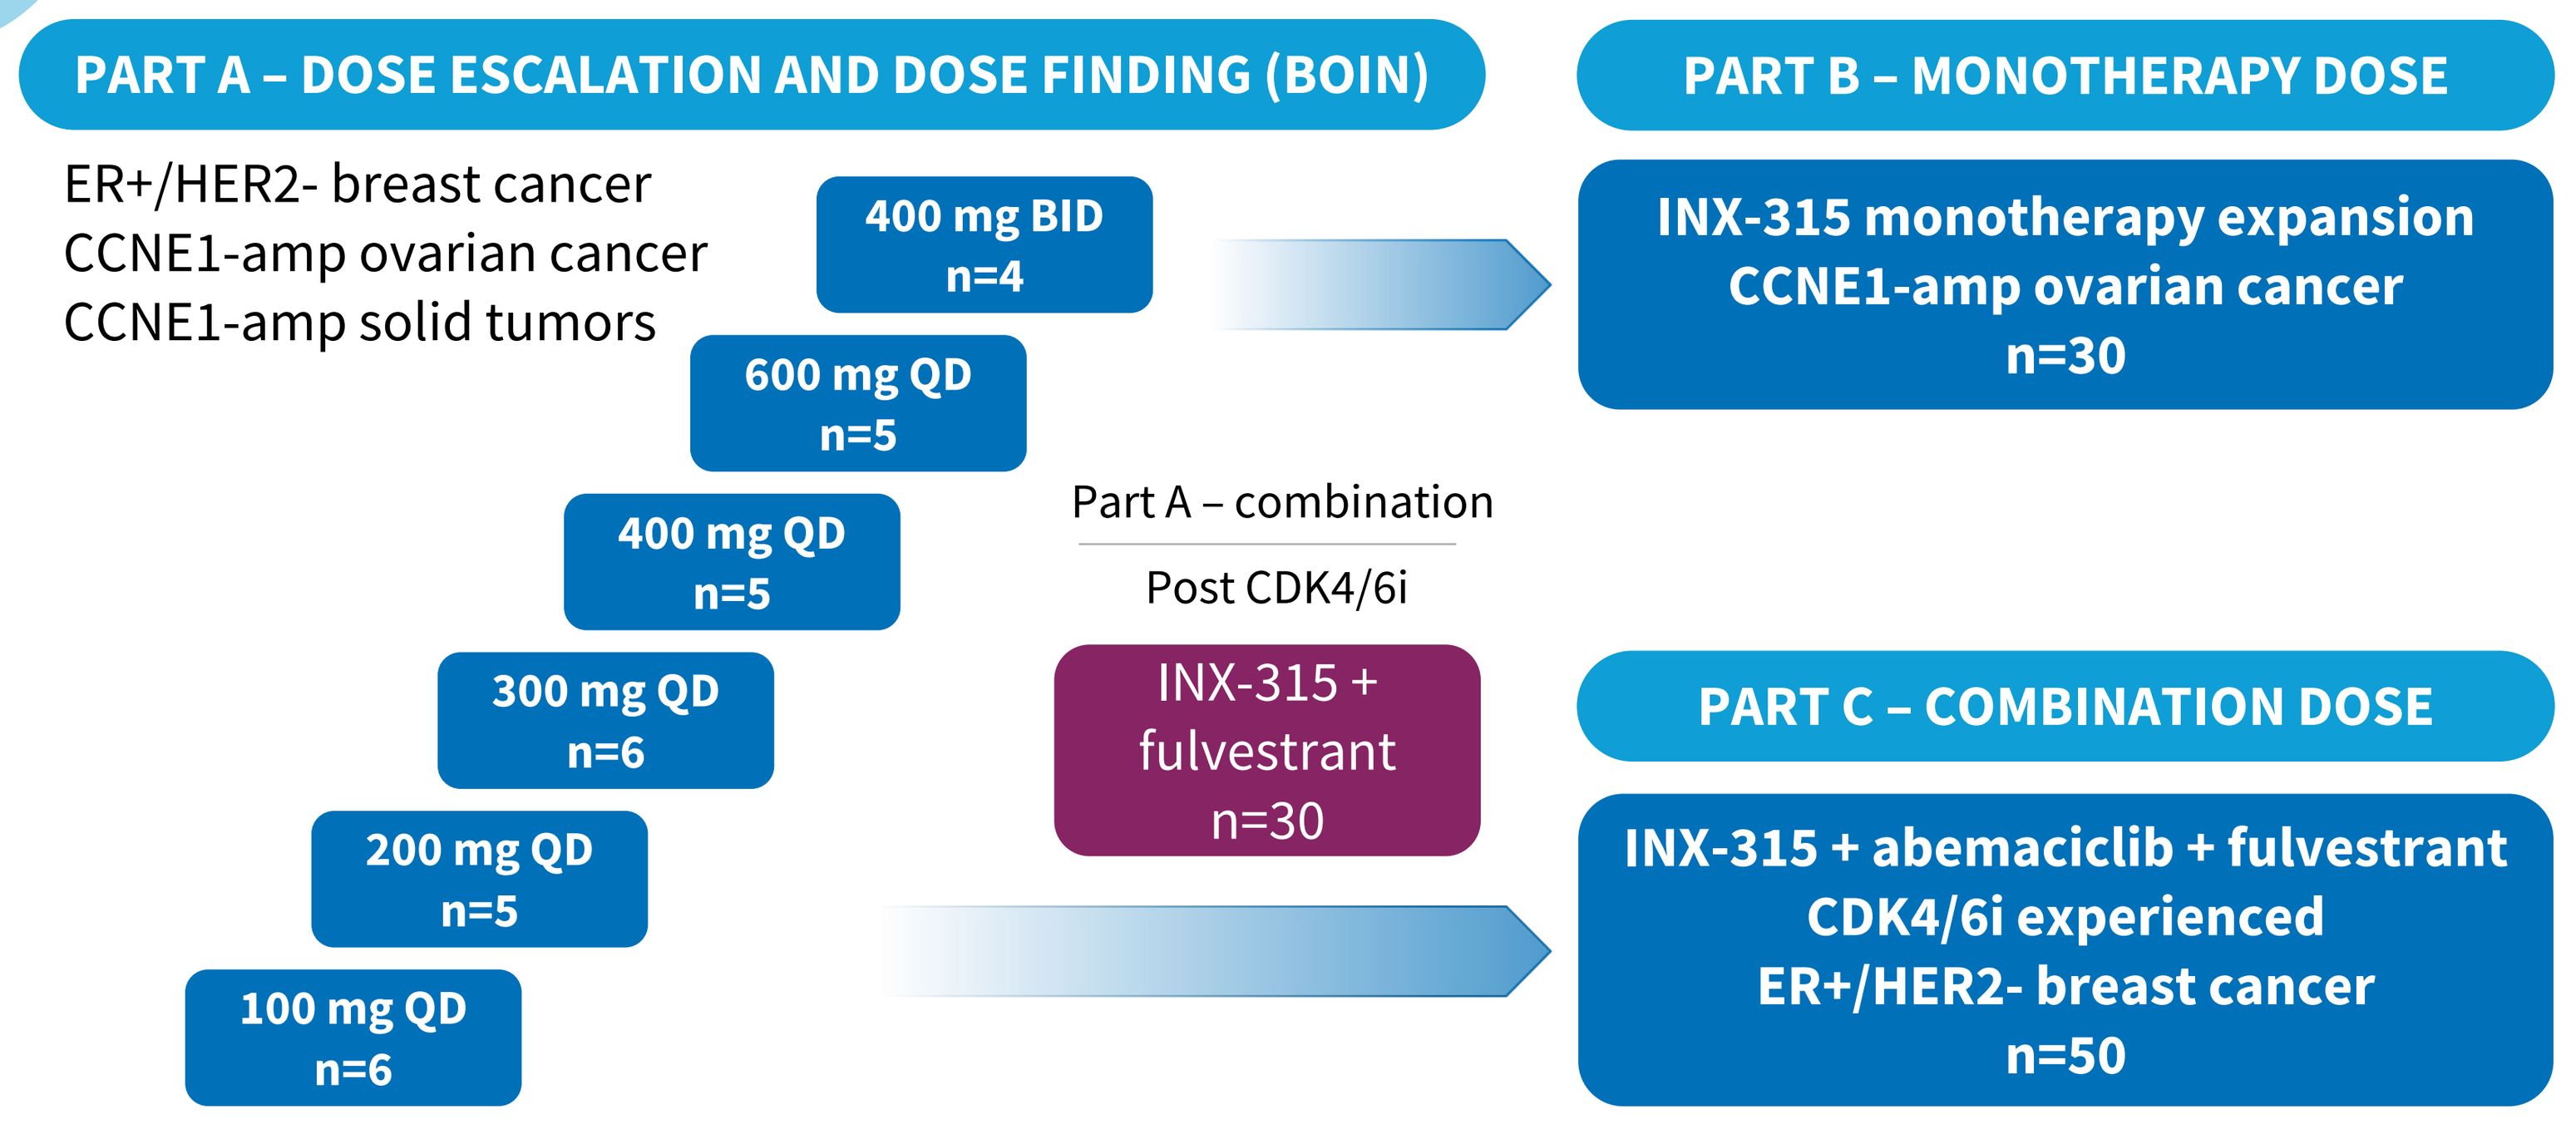
PART A – DOSE ESCALATION AND DOSE FINDING (BOIN)
ER+/HER2- breast cancer
CCNE1-amp ovarian cancer
CCNE1-amp solid tumors
400 mg BID n=4
600 mg QD n=5
400 mg QD n=5
300 mg QD n=6
200 mg QD n=5
100 mg QD n=6
Part A – combination
Post CDK4/6i
INX-315 + fulvestrant n=30
PART B – MONOTHERAPY DOSE
INX-315 monotherapy expansion
CCNE1-amp ovarian cancer
n=30
PART C – COMBINATION DOSE
INX-315 + abemaciclib + fulvestrant
CDK4/6i experienced
ER+/HER2- breast cancer
n=50

Figure 1. Study schema

PHARMACOKINETICS

- INX-315 plasma concentrations increased in a dose proportional manner
- Steady-state trough concentrations were ≥ the predicted effective concentration of 100 ng/mL
- Low Cmax to Cmin ratio likely to translate to best-in-class safety

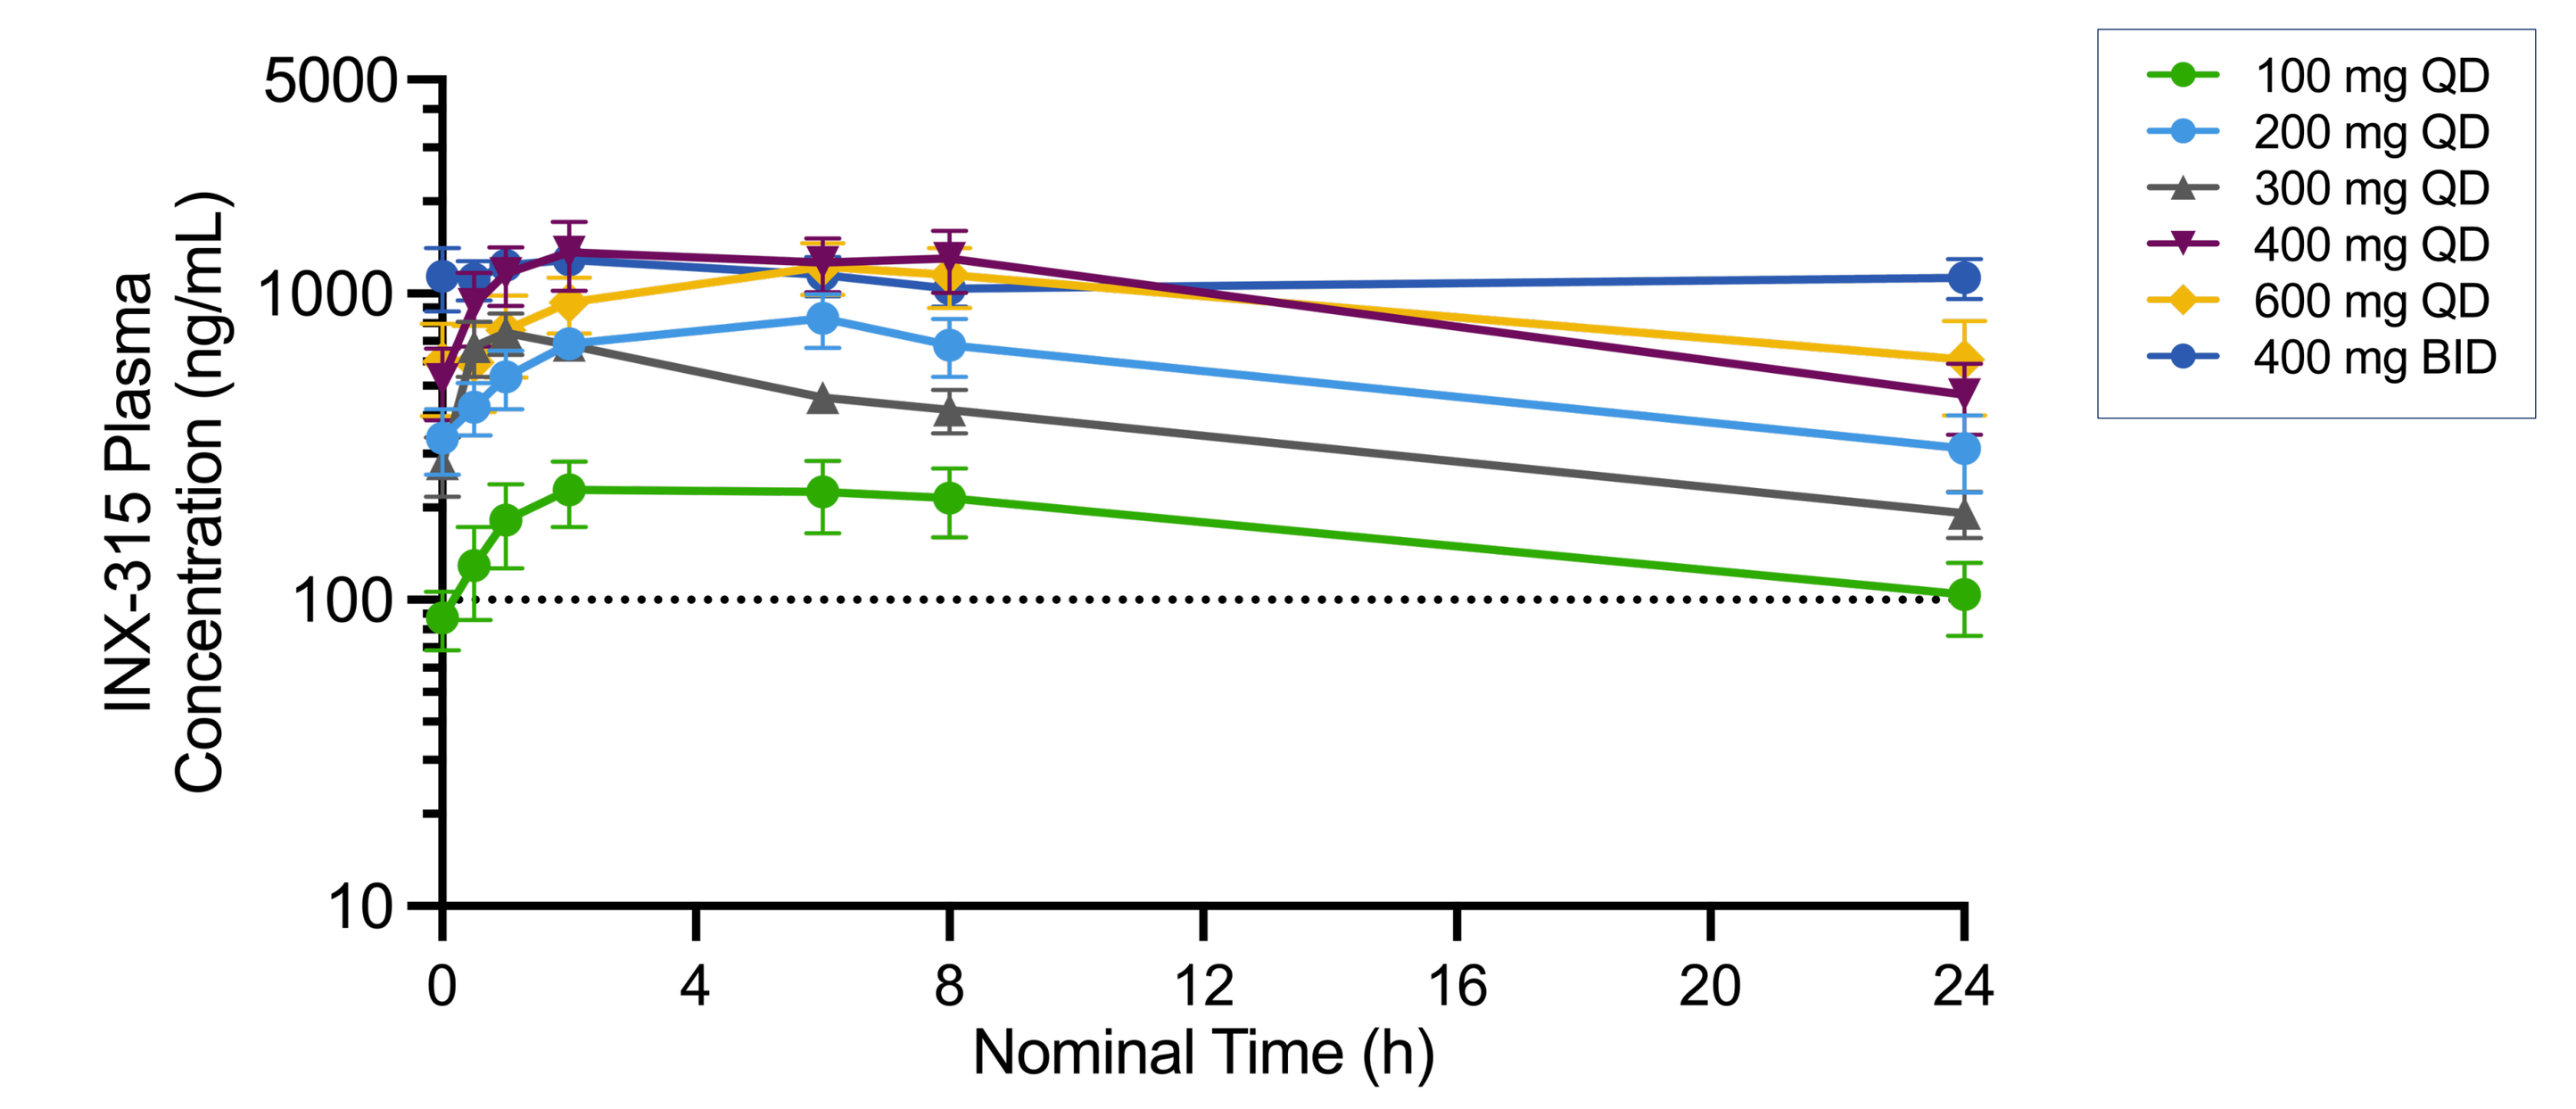
5000
1000
100
10
INX-315 Plasma Concentration (ng/mL)
0
4
8
12
16
20
24
Nominal Time (h)
100 mg QD
200 mg QD
300 mg QD
400 mg QD
600 mg QD
400 mg BID

Figure 2. Day 15 mean INX-315 plasma concentrations by cohort

TABLE 1. DEMOGRAPHICS AND BASELINE CHARACTERISTICS, PART A

CHARACTERISTIC	PATIENTS (N=31)
CHARACTERISTIC	
Age (years), median (range)	60 (29, 78)
Sex, N (%)	
Male	8 (26)
Female	23 (74)
Race, N (%)	
White	24 (77)
Asian	4 (13)
Black or African American	2 (7)
Other or not reported	1 (3)
Tumor type, N (%)	
ER+/HER2- breast cancer	10 (32)
HGSOC/fallopian (CCNE1 amplified)	10 (32)
Other solid tumors (CCNE1 amplified)	11(36)
Baseline ECOG, N (%)	
0	19 (61)
1	12 (39)
Prior lines, median (range)	4 (1, 9)
BREAST CANCER PATIENTS ONLY (N=10)	
Prior CDK4/6 inhibitor, n (%)	10 (100)
Prior fulvestrant, n (%)	9 (90)
Prior aromatase inhibitor, n (%)	9 (90)
Prior chemotherapy, n (%)	9 (90)
Prior antibody drug conjugate, n (%)	3 (30)

SAFETY

- Treatment-related adverse events (TRAEs) were observed in 26 patients (83.9%) (Table 2)
- The most frequent treatment-related AEs were thrombocytopenia (48.4%), nausea (38.7%), neutropenia (38.7%), and diarrhea (29.0%)
- Serious adverse events (SAEs) occurred in 10 (32.2%) patients, none were considered related to INX-315
- No fatal AEs were reported, and no AEs led to treatment discontinuation
- Dose limiting toxicity (DLT) was observed in 1 patient (grade 3 fatigue at 600 mg QD) resulting in a dose reduction
- Evaluation of the 400 mg BID dose level is ongoing
- 600 mg INX-315 QD has been selected by the safety monitoring committee for combination with fulvestrant and monotherapy expansion in Part B

TABLE 2. TRAEs OCCURRING IN ≥15% OF PATIENTS BY GRADE (PART A, SAFETY EVALUABLE PATIENTS, N=31)

TRAE BY PT, N(%)	GRADE 1	GRADE 2	GRADE 3	GRADE 4	TOTAL
With ≥1 TRAE	9 (29.0)	12 (38.7)	4 (12.9)	1 (3.2)	26 (83.9)
Thrombocytopenia	11 (35.5)	3 (9.7)	0	1 (3.2)	15 (48.4)
Nausea	9 (29.0)	3 (9.7)	0	0	12 (38.7)
Neutropenia	2 (6.5)	6 (19.4)	3 (9.7)	1 (3.2)	12 (38.7)
Diarrhea	6 (19.4)	2 (6.5)	1 (3.2)	0	9 (29.0)
Anemia	4 (12.9)	2 (6.5)	1 (3.2)	0	7 (22.6)
Vomiting	5 (16.1)	1 (3.2)	0	0	6 (19.4)
Fatigue	1 (3.2)	2 (6.5)	2 (6.5)	0	5 (16.1)
Leukopenia	1 (3.2)	2 (6.5)	2 (6.5)	0	5 (16.1)

EFFICACY

- Among all response-evaluable patients in Part A (n=30), 3 (10%) had a partial response and 19 (63%) had stable disease (Table 3; Figure 3)
- In patients with ER+/HER2- breast cancer, PR was observed in 1 (10%) and SD was observed in 5 (50%)
- In patients with CCNE1 amplified HGSOC/fallopian, PR was observed in 2 (20%) and SD was observed in 8 (80%)
- Pharmacodynamic studies (ctDNA, TK activity) are ongoing

TABLE 3. BEST OVERALL RESPONSE PER INVESTIGATOR ASSESSMENT

N (%)	ALL PATIENTS (N=30)	BREAST CANCER (N=10)
Complete response (CR)	0	0
Partial response (PR)	3 (10)	1 (10)
Stable disease (SD)	19 (63)	5 (50)
Progressive disease (PD)	8 (27)	4 (40)

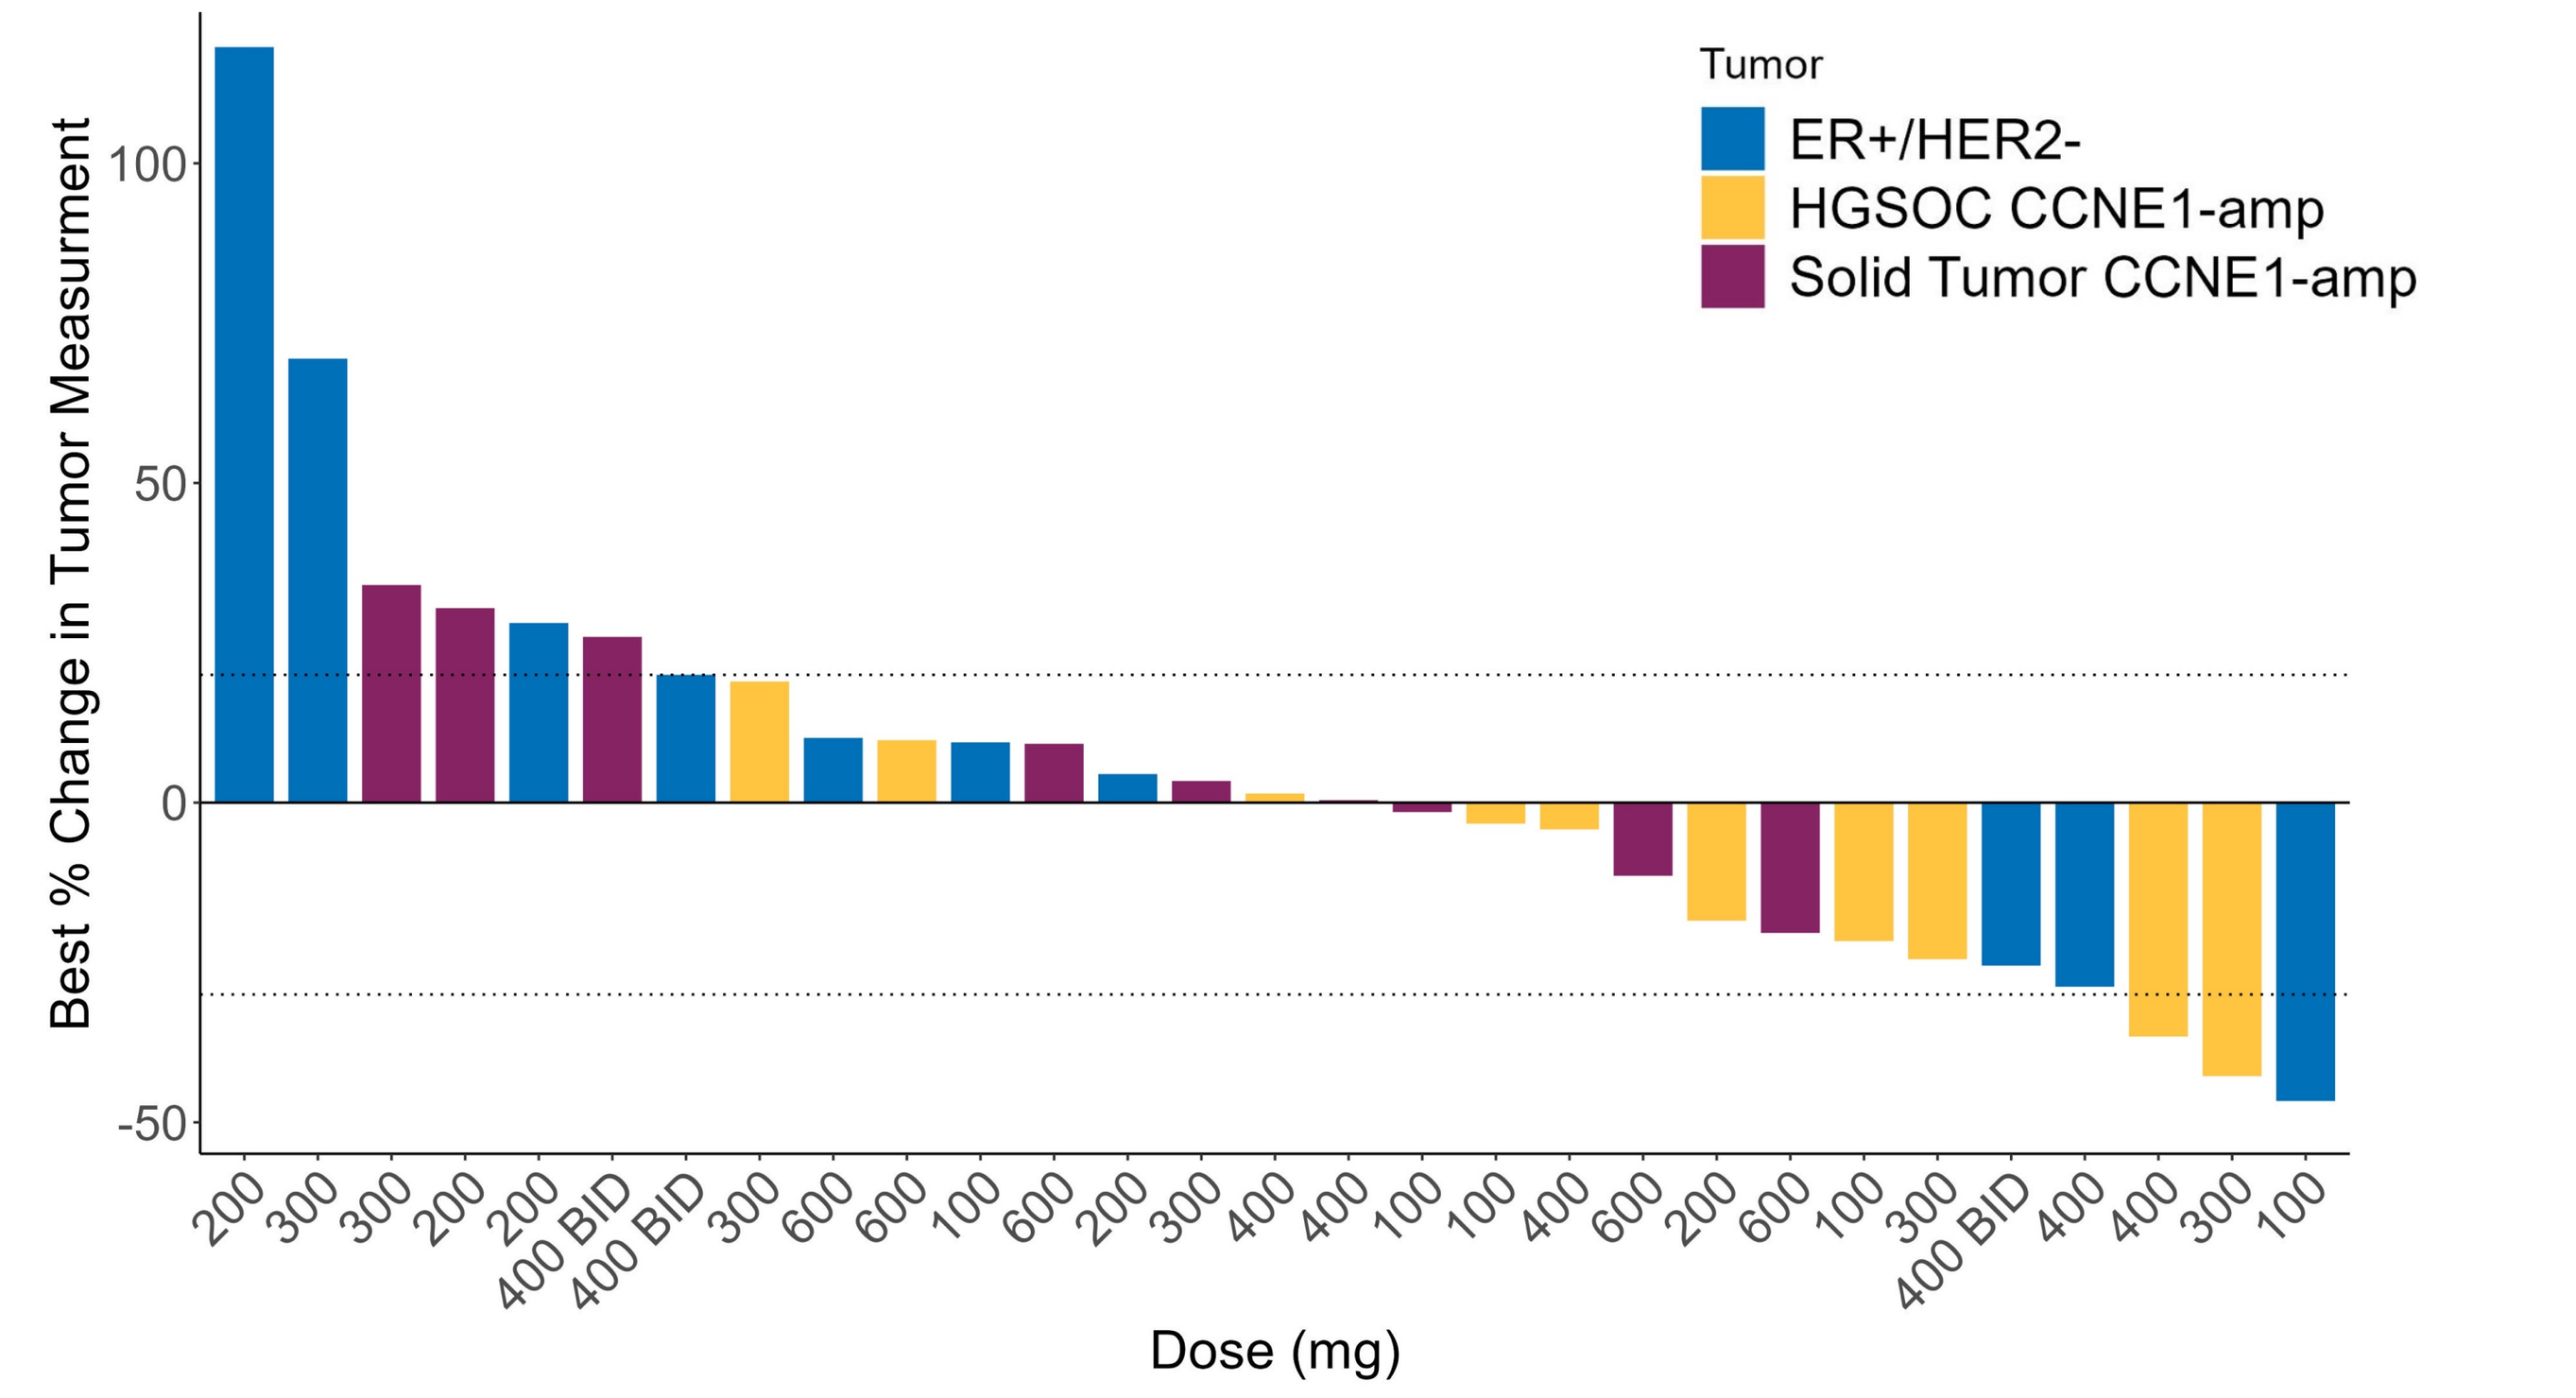
Tumor
ER+/HER2-
HGSOC CCNE1-amp
Solid Tumor CCNE1-amp
Best % Change in Tumor Measurment
100
50
0
-50
200 300 300 200 200 400 BID 400 BID 300 600 600 100 600 200 300 400 400 100 100 400 600 200 600 100 300 400 BID 400 400 300 100
Dose (mg)

Figure 3. Waterfall plot of best percent change from baseline in sum of diameters for all patients (RECIST v1.1) (N=29)

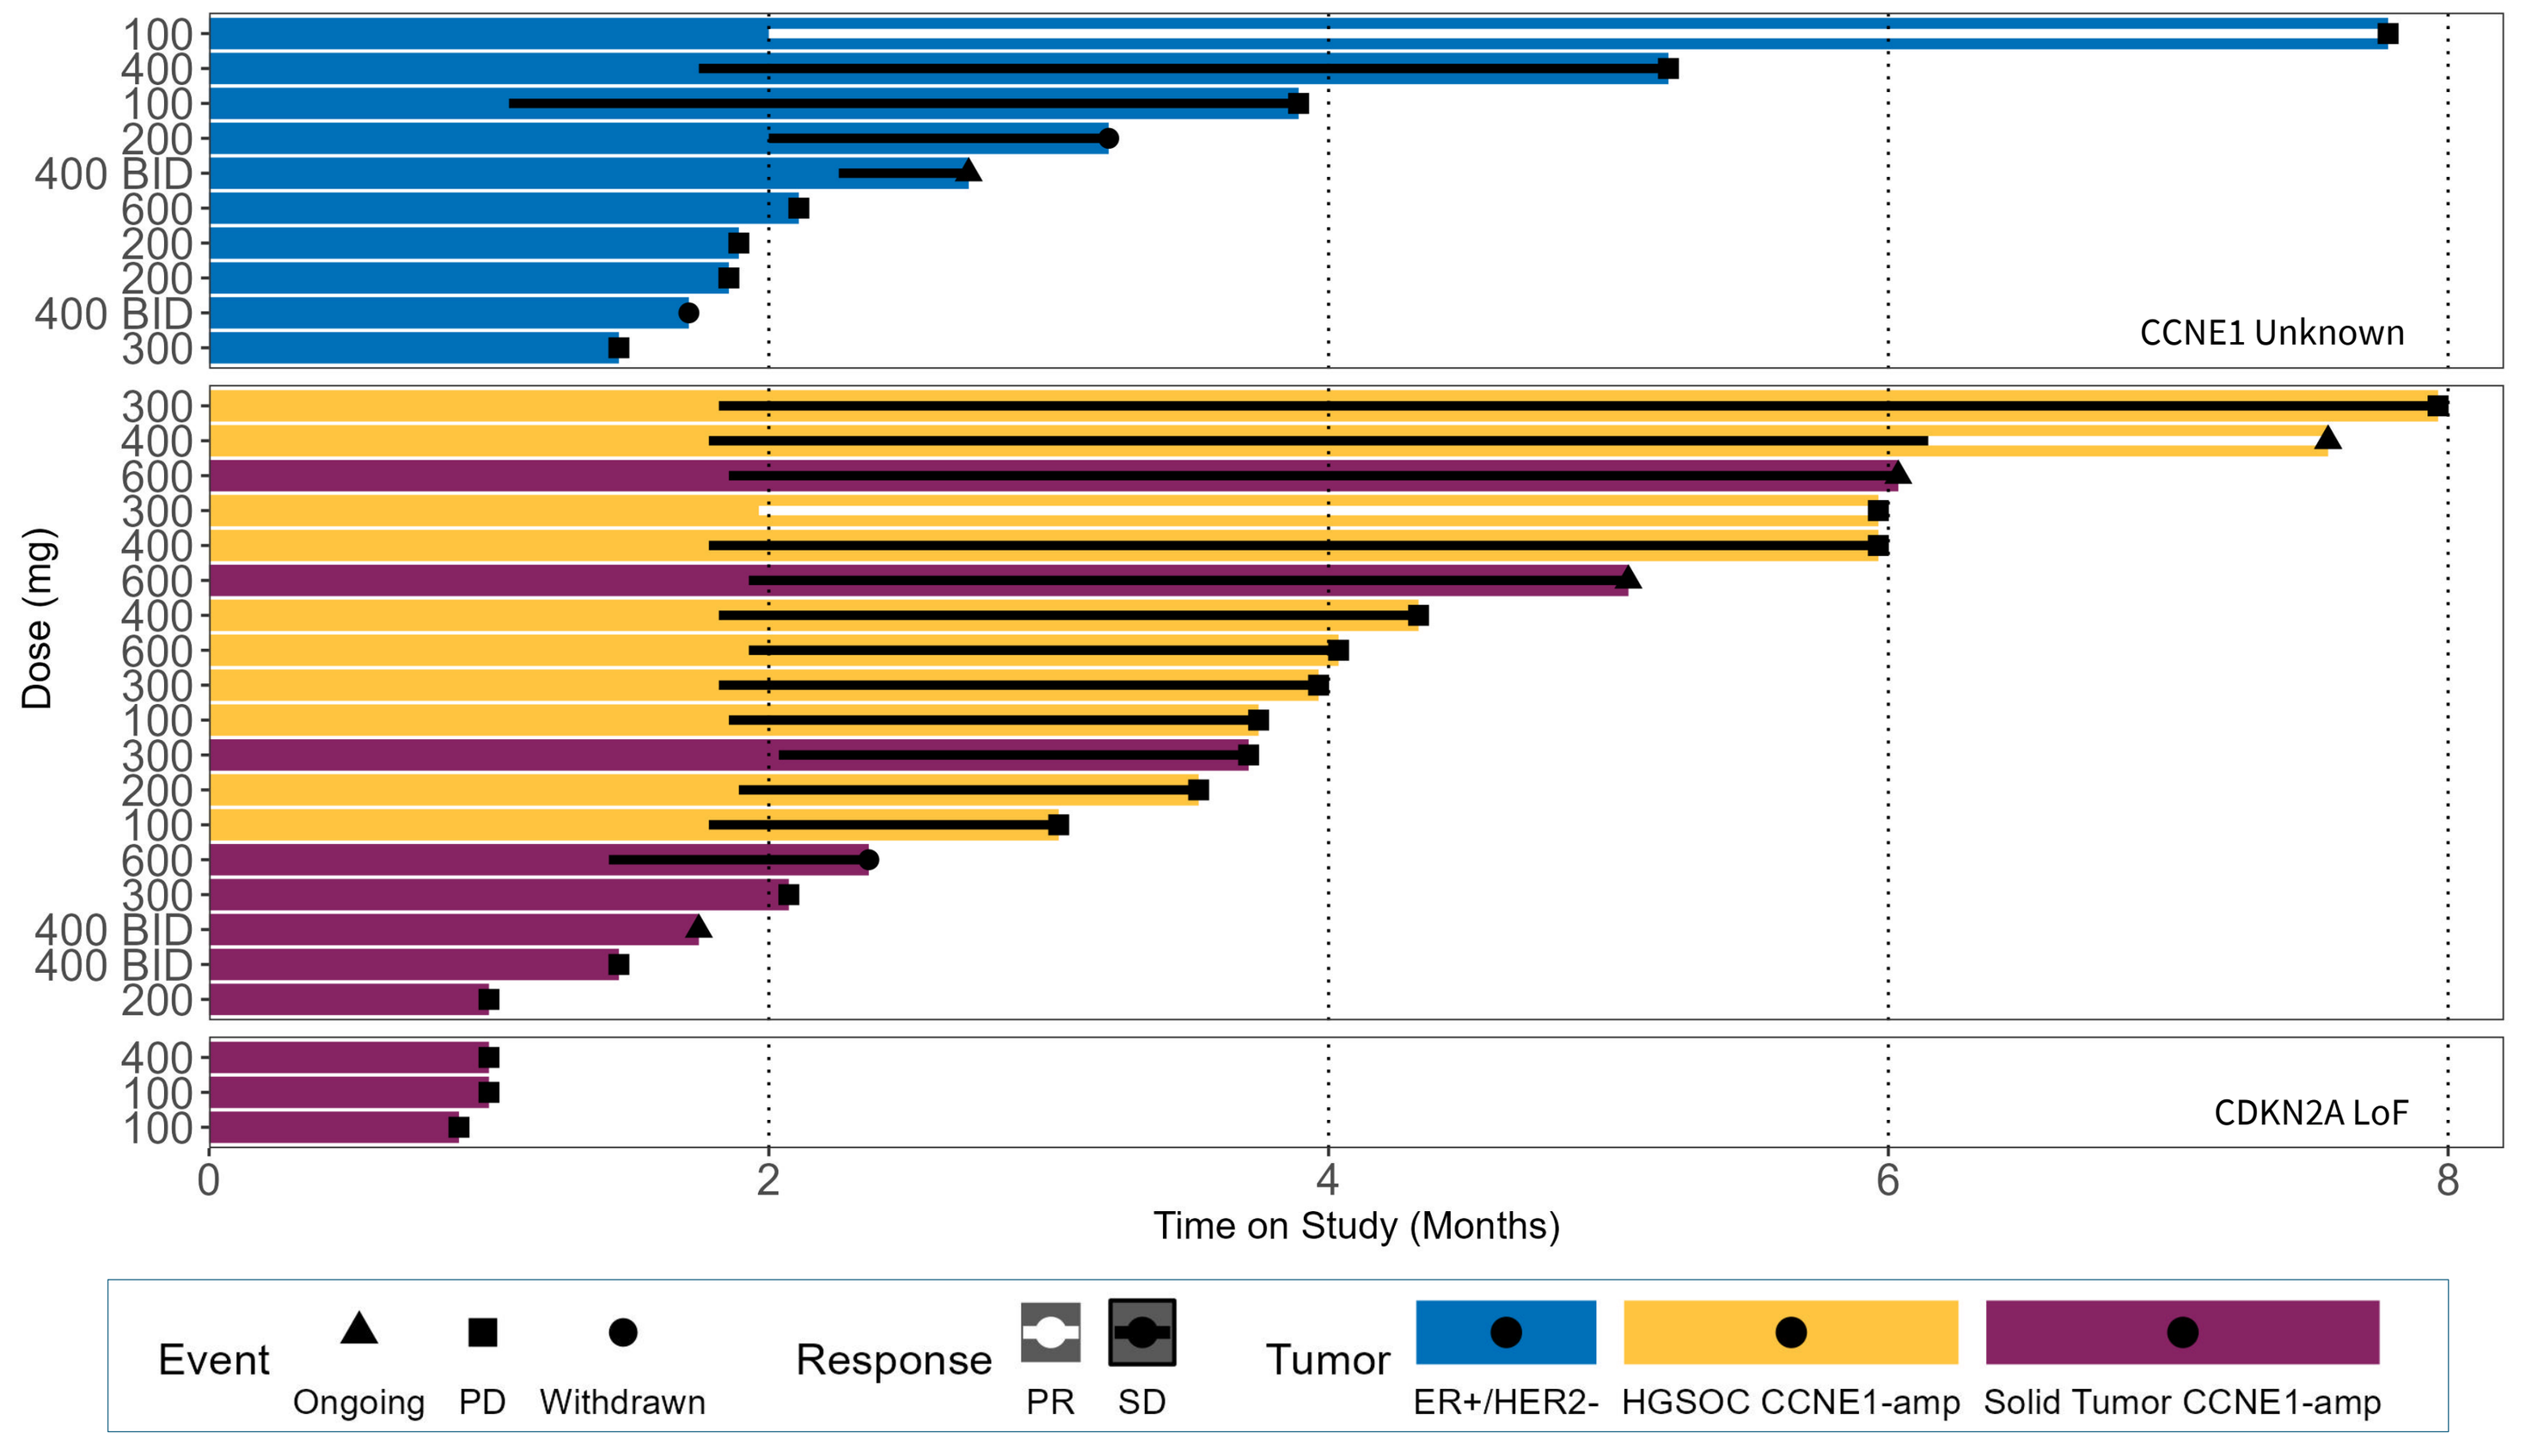
Dose (mg)
100 400 100 200 400 BID 600 200 200 400 BID 300
CCNE1 Unknown
300 400 600 300 400 600 400 600 300 100 300 200 100 600 300 400 BID 400 BID 200
400 100 100
CDKN2A LoF
0
2
4
6
8
Time on Study (Months)
Event
Ongoing
PD
Withdrawn
Response
PR
SD
Tumor
ER+/HER2-
HGSOC CCNE1-amp
Solid Tumor CCNE1-amp

Figure 4. Swimmer plot (RECIST v1.1) (N=31)

ACKNOWLEDGEMENTS:
- We thank all the investigators and site staff, with special thanks to the patients and their families
- We thank Incyclix Bio team members who supported the creation of this poster
- Study sponsored by Incyclix Bio, LLC
- Editorial assistance was provided by Molly Jenkins, funded by Incyclix Bio, LLC

DISCLAIMER:
This presentation is the intellectual property of the author/presenter. Copies of this poster are for personal use only and may not be reproduced without permission from SABCS® and the author of this poster. Authors may be contacted at Antoinette.Tan@atriumhealth.org or proberts@incyclixbio.com for permission to reprint and/or distribute.